Evaluation of Progression Free Survival as a Surrogate for Overall Survival for prostate cancer treatment: Software development

Theophile Bigirumurame[1], Ziv Shkedy[1], Tomasz Burzykowski[1]

Joris Dries[2], Suzy Van Sanden[2], Pushpike Thilakarathne [2]

[1] Interuniversity Institute for Biostatistics, Hasselt University, Belgium, [2]Janssen, Beerse, Belgium

INTRODUCTION

In clinical trials, the determination of the true endpoint or the effect of a new therapy on the true endpoint may be difficult, requiring an expensive, invasive or uncomfortable procedure. In some trials, however, the main endpoint of interest (true endpoint), for example death, is rare and/or takes a long time to reach. In such trials, there would be benefit in finding a more proximate endpoint (surrogate endpoint) to determine more quickly the effect of an intervention [1].

The validation and evaluation of a surrogate endpoint needs to be assessed at both the patient and the trial-level [2]. On one hand, the individual-level surrogacy measures the association between the potential surrogate endpoint and the true endpoint after adjusting for treatment effects on both true and surrogate endpoints.

On the other hand, the trial-level surrogacy describes how well one can predict the treatment effect on the true endpoint in a future trial based on the observed association between the treatment effects on the surrogate and true endpoints observed in previous trials [3].

Unfortunately, standard software to perform analysis to validate surrogate endpoints is lacking. To that end, we developed a user friendly SAS macro to automate such an analysis. We demonstrate the usage and capacities of the software implementation using a clinical trial dataset with two failure time endpoints.

METHODS

In validation of a possible surrogate, two major branches have been developed, methods in single-trial settings and meta-analytical evaluation methods. The second one has been applied in our analysis.

Firstly, a bivariate copula model was used to measure association between failure time endpoints. It was assumed that the joint survival function for (S_{ij}, T_{ij}) could be written as:

$$F(s,t)=P(S_{ij}\geq s,T_{ij}\geq t)=C_{\theta}\{F_{S_{ij}}(s),F_{T_{ij}}(t)\} \qquad s,t\geq 0,$$

where F_{Sij} and F_{Tij} denote the marginal survival functions for both endpoints [3]. C_{θ} is a copula, a bivariate distribution function on $[0,1]^2$. The marginal distribution were given by the following equations:

$$F_{S_{ij}}(s)=\exp\{-\int_0^s \lambda_{Si}(x)\exp(\alpha_i Z_{ij})dx\},$$

$$F_{T_{ij}}(t)=\exp\{-\int_0^t \lambda_{Ti}(x)\exp(\beta_i Z_{ij})dx\},$$

where λ_{Si} and λ_{Ti} are trial specific marginal baseline hazard function, and α_i, β_i are trial specific effects of treatment Z on the endpoint in trial i.

At the second stage it is assumed that:

$$\begin{pmatrix}\alpha_i\\ \beta_i\end{pmatrix}=\begin{pmatrix}\alpha\\ \beta\end{pmatrix}+\begin{pmatrix}a_i\\ b_i\end{pmatrix},$$

where the second term on the right hand side is assumed to follow a zero mean normal distribution with dispersion matrix:

$$D_r=\begin{pmatrix}d_{aa} & d_{ab}\\ & d_{bb}\end{pmatrix}.$$

Trial level surrogacy is assessed based on the coefficient of determination:

$$R^2_{trial}=\frac{d^2_{ab}}{d_{aa}d_{bb}} .$$

Individual level surrogacy is assessed based on Kendall's τ, as it depends only on the copula function but not the marginal distribution of S_{ij} and T_{ij}.

$$\tau=4\int_0^1\int_0^1 C_{\theta}(F_{S_{ij}},F_{T_{ij}})C_{\theta}(dF_S,dF_T)-1$$

Unfortunately, there is no standard software to perform this validation. A SAS macro has been developed to perform the analysis.

SAS MACRO CALL

```
%macro cont(data=,true=,trueind=,surr=,surrind=,trt=, center=);

Data: is the dataset containing one record for each subject, with
      the following variables:
True: variable representing the true endpoint,
Trueind: censoring indicator (1=event, 0= censoring),
Surr: variable containing the surrogate endpoint,
Surrind: censoring indicator (1=event, 0=censoring),
Trt: treatment indicator (0 or 1),
Center: unique id (continuous) for units for which treatment
effects are estimated,
Trial: unique id (continuous) for the group of unit, for which
common baselines are to be used,
```

Dataset

The data that are analyzed come from a multicenter trial conducted in 13 countries.
Patients were randomized to an active treatment and on a control treatment. Some centers were having only patients on one treatment arm. The true endpoint was overall survival and the surrogate was progression free survival. The median survival time was 485 days on OS and 88 days on PFS for active group (340 and 85 respectively for control group). Figure 1 shows the Kaplan Meier survival curves .

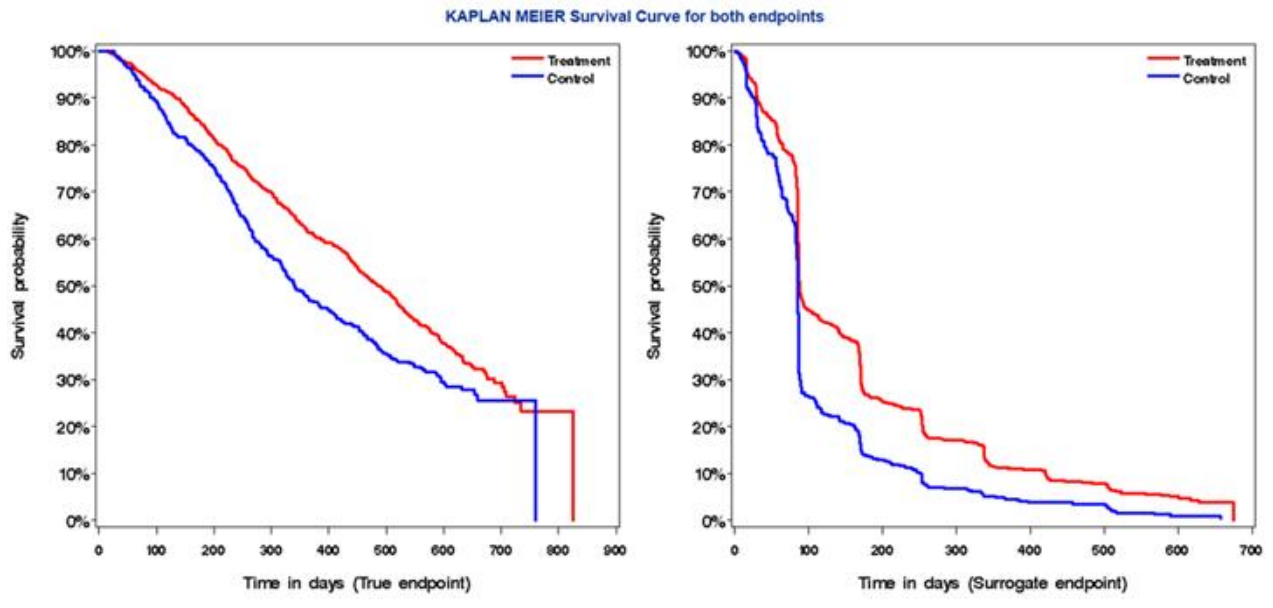
KAPLAN MEIER Survival Curve for both endpoints
Treatment
Control
Survival probability
Time in days (True endpoint)
Time in days (Surrogate endpoint)

Fig1: Kaplan Meier survival curves for both endpoints stratified by treatment arm

RESULTS

A trial level surrogacy equal to 0.31 and individual level surrogacy equal to 0.3 were obtained. Figure 2 shows the estimated treatment effect on the true endpoint vs treatment effect obtained on the surrogate from the developed macro. Table 1 summarizes obtained estimates with their confidence interval.

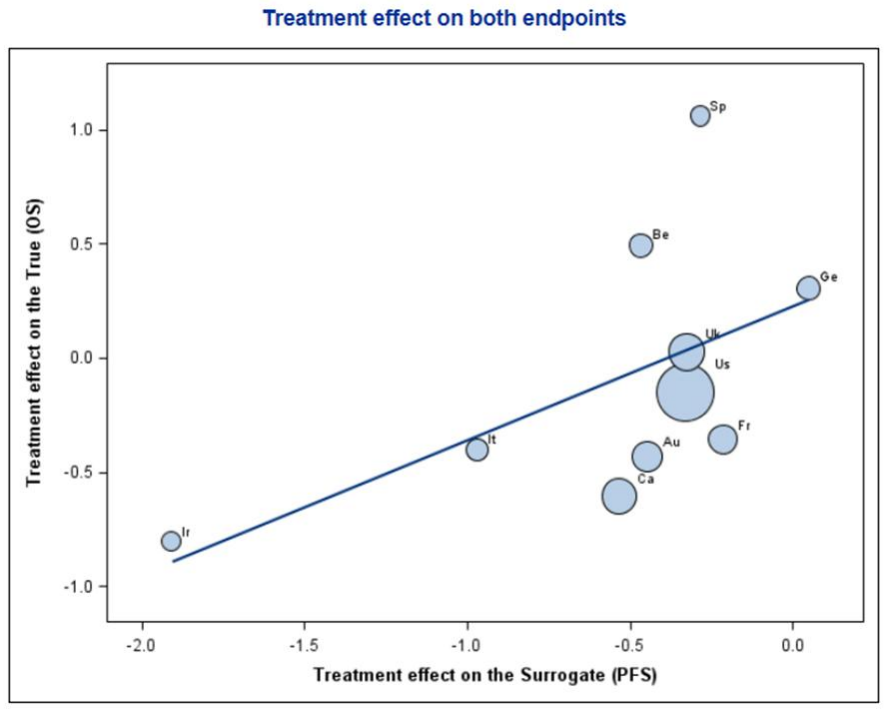
Treatment effect on both endpoints
Treatment effect on the True (OS)
Treatment effect on the Surrogate (PFS)
Sp
Be
Ge
Uk
Us
Fr
It
Au
Ca
Ir

Fig2: Treatment effect on overall survival versus Treatment effect on progression free survival (left panel). Point size proportional to the number of subjects in each center. The straight line contains predictions from a weighted linear regression.

SURROGACY MEASURES

INDIVIDUAL			TRIAL		
LOWER	TAU	UPPER	LOWER	R square	UPPER
0.2635	0.3039	0.3442	-0.1592	0.3175	0.7942

Table1: Estimated individual level and trial level surrogacy measures

DISCUSSION AND CONCLUSIONS

- The obtained individual and trial level surrogacy values show relatively low association between overall survival and progression free survival.
- For a good surrogate, obtained estimates should be close to one, with short confidence intervals.
- For this dataset progression free survival may not be a good surrogate for overall survival.

REFERENCES

[1] Ellenbergh,S.S and Hamilton, J.M (1989) Surrogate endpoints in clinical trials:cancer. *Statistics in Medecine*, **8**,405-413.
[2] Bruzykowski, T. , Molenberghs, G., Buyse, M., Renard, D., and Geys, H. (2001) Validation of surrogate endpoints in multiple randomized clinical trials with failure-time endpoints. *Applied statistics* , **50**, 405-422.
[3] Bruzykowski, T., Molenberghs, G., and Buyse, M. *The evaluation of surrogate endpoints*. New York: Springer-Verlag (2005).